P779

Tetra-specific MATCH4 antibodies mediate concomitant blockade of PD-L1 and potent T cell-stimulation, locally restricted to Her2$^+$ tumors

Alexandre Simonin[1], Tea Gunde[1], Christian Hess[1], Stefan Warmuth[1], Matthias Brock[1], Dan Snell[1], Julia Tietz[1], Eva Oswald[2], Robin Heiz[1], Naomi Flückiger[1], Julia Zeberer[1], Dania Diem[1], Dana Mahler[1], Diego Morenzoni[1], Simone Muntwiler[1], Benjamin Küttner[1], Sebastian Meyer[1], Timothy Egan[3], David Urech[1]

1) Numab Therapeutics AG, Einsiedlerstrasse 34, 8820 Wädenswil, Switzerland; 2) Charles River Discovery Service Germany GmbH, Am Flughafen 12-14, 79108 Freiburg, Germany; 3) Numab US LLC, P.O. Box 902 Northboro, 01532 Massachusetts, US

Tumor-targeted T cell activation mechanisms have proved highly effective in the redirection of the immune system towards cancer cells. Of particular note is the use of CD3 T cell engagers and the approval of the molecule Blincyto for the treatment of patients suffering from B-ALL. One issue in identifying appropriate tumor associated antigens (TAAs) for CD3 T cell engagement is ensuring an appropriate expression level in tumor tissue vs normal tissue to preferentially drive T cell mediated cytotoxicity to the tumor.

We sought to address this issue and open the therapeutic window of CD3 engagers by the generation of an affinity tailored MATCH4 molecule targeting PD-L1, Her2, CD3 and human serum albumin (hSA) to preferentially mediate T cell killing of tumor cells.

We demonstrate that PD-L1/PD-1 antagonism is retained within this molecule which has increased potency compared to a conventional CD3 x Her2 engager, in a single-chain diabody format (scDb), on both Her2 high as well as Her2 low cells, in the presence of simultaneous PD-L1 expression. The activity is not enhanced on PDL1-negative cells and therefore we demonstrate the potential to open the therapeutic window towards higher activity on cancer cells in the context of PD-L1 expression.

Concept

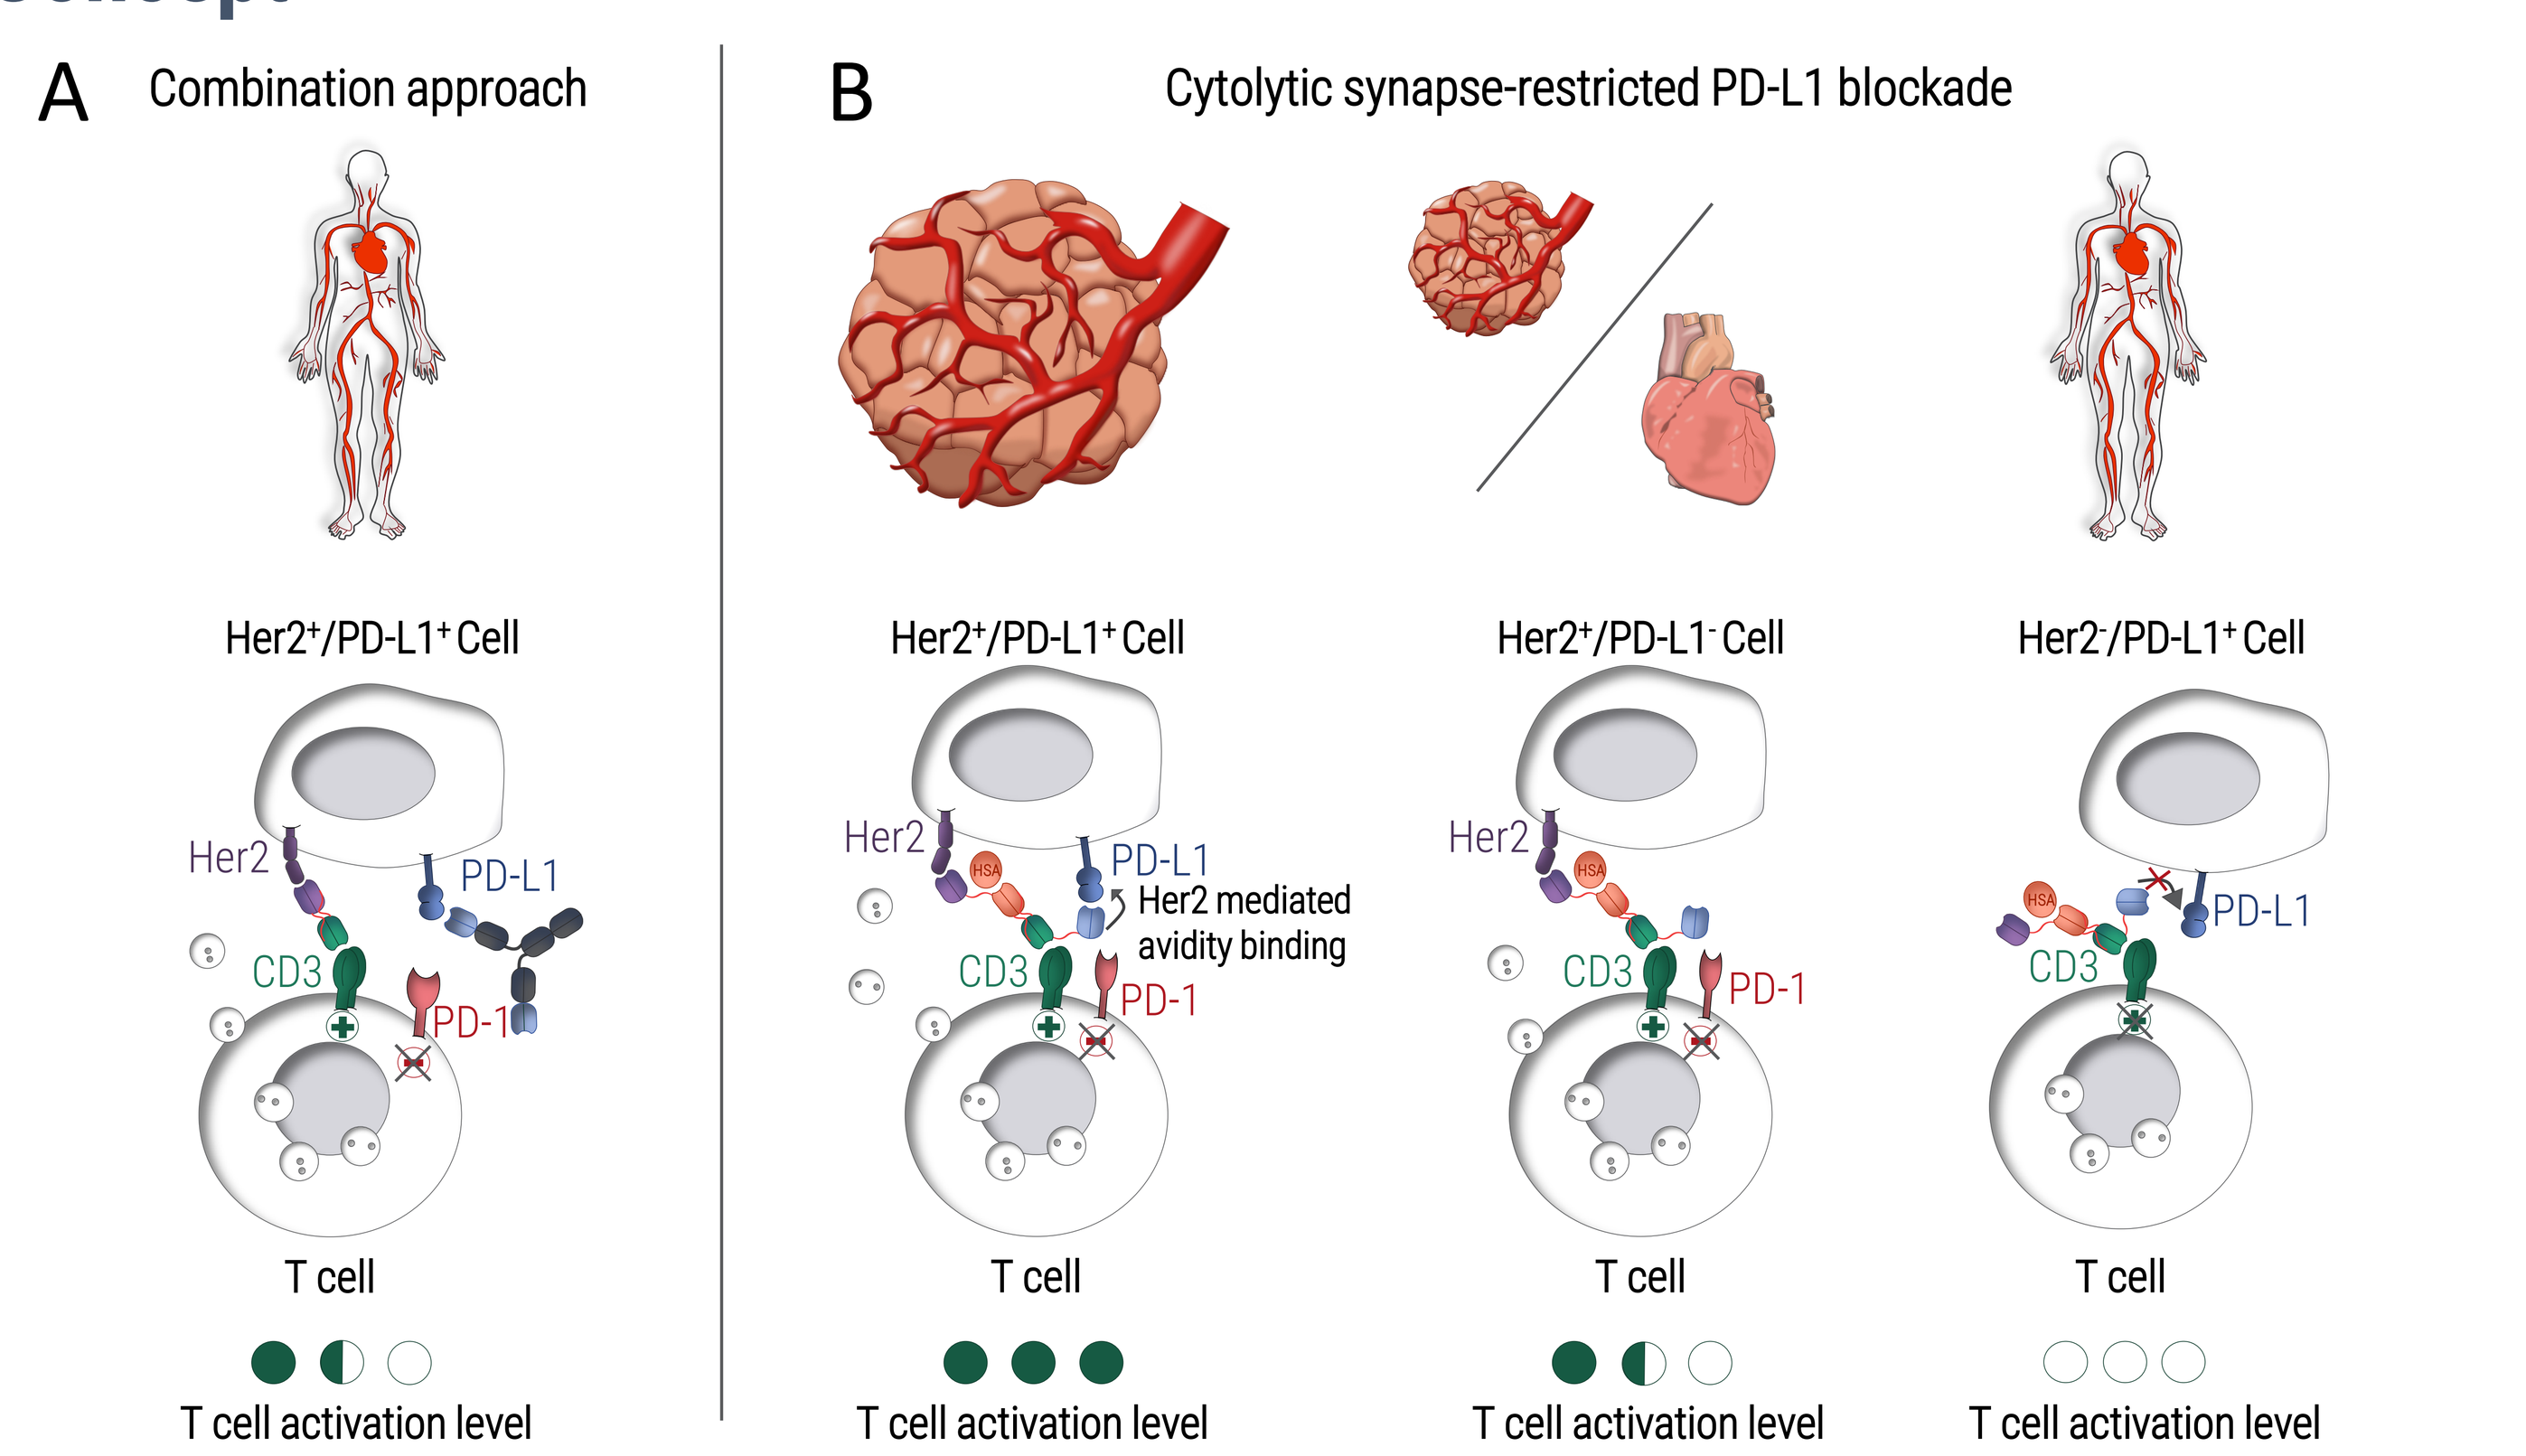

A Combination approach
B Cytolytic synapse-restricted PD-L1 blockade
Her2+/PD-L1+ Cell
Her2+/PD-L1+ Cell
Her2+/PD-L1- Cell
Her2-/PD-L1+ Cell
Her2
PD-L1
CD3
PD-1
Her2 mediated avidity binding
T cell
T cell activation level

Figure 1. A) Conventional combination of a Her2/CD3 bispecific molecule with PD-L1 blockade results in potent T cell-activation and target cell lysis. **B)** αHer2-/αPD-L1-mediated avidity-binding elicits more potent T cell-activation and target cell lysis than the combination therapy in the presence of Her2$^+$PD-L1$^+$ cancer cells, of both Her2-high and Her2-low phenotypes. Targeting of Her2$^+$PD-L1$^-$ cells is similarly effective as combination therapy, whilst importantly Her2$^-$PD-L1$^+$ cells are spared from cytolysis.

Affinity-optimized anti-PD-L1 domain specifically targeting Her2/PD-L1 expressing cancer cells

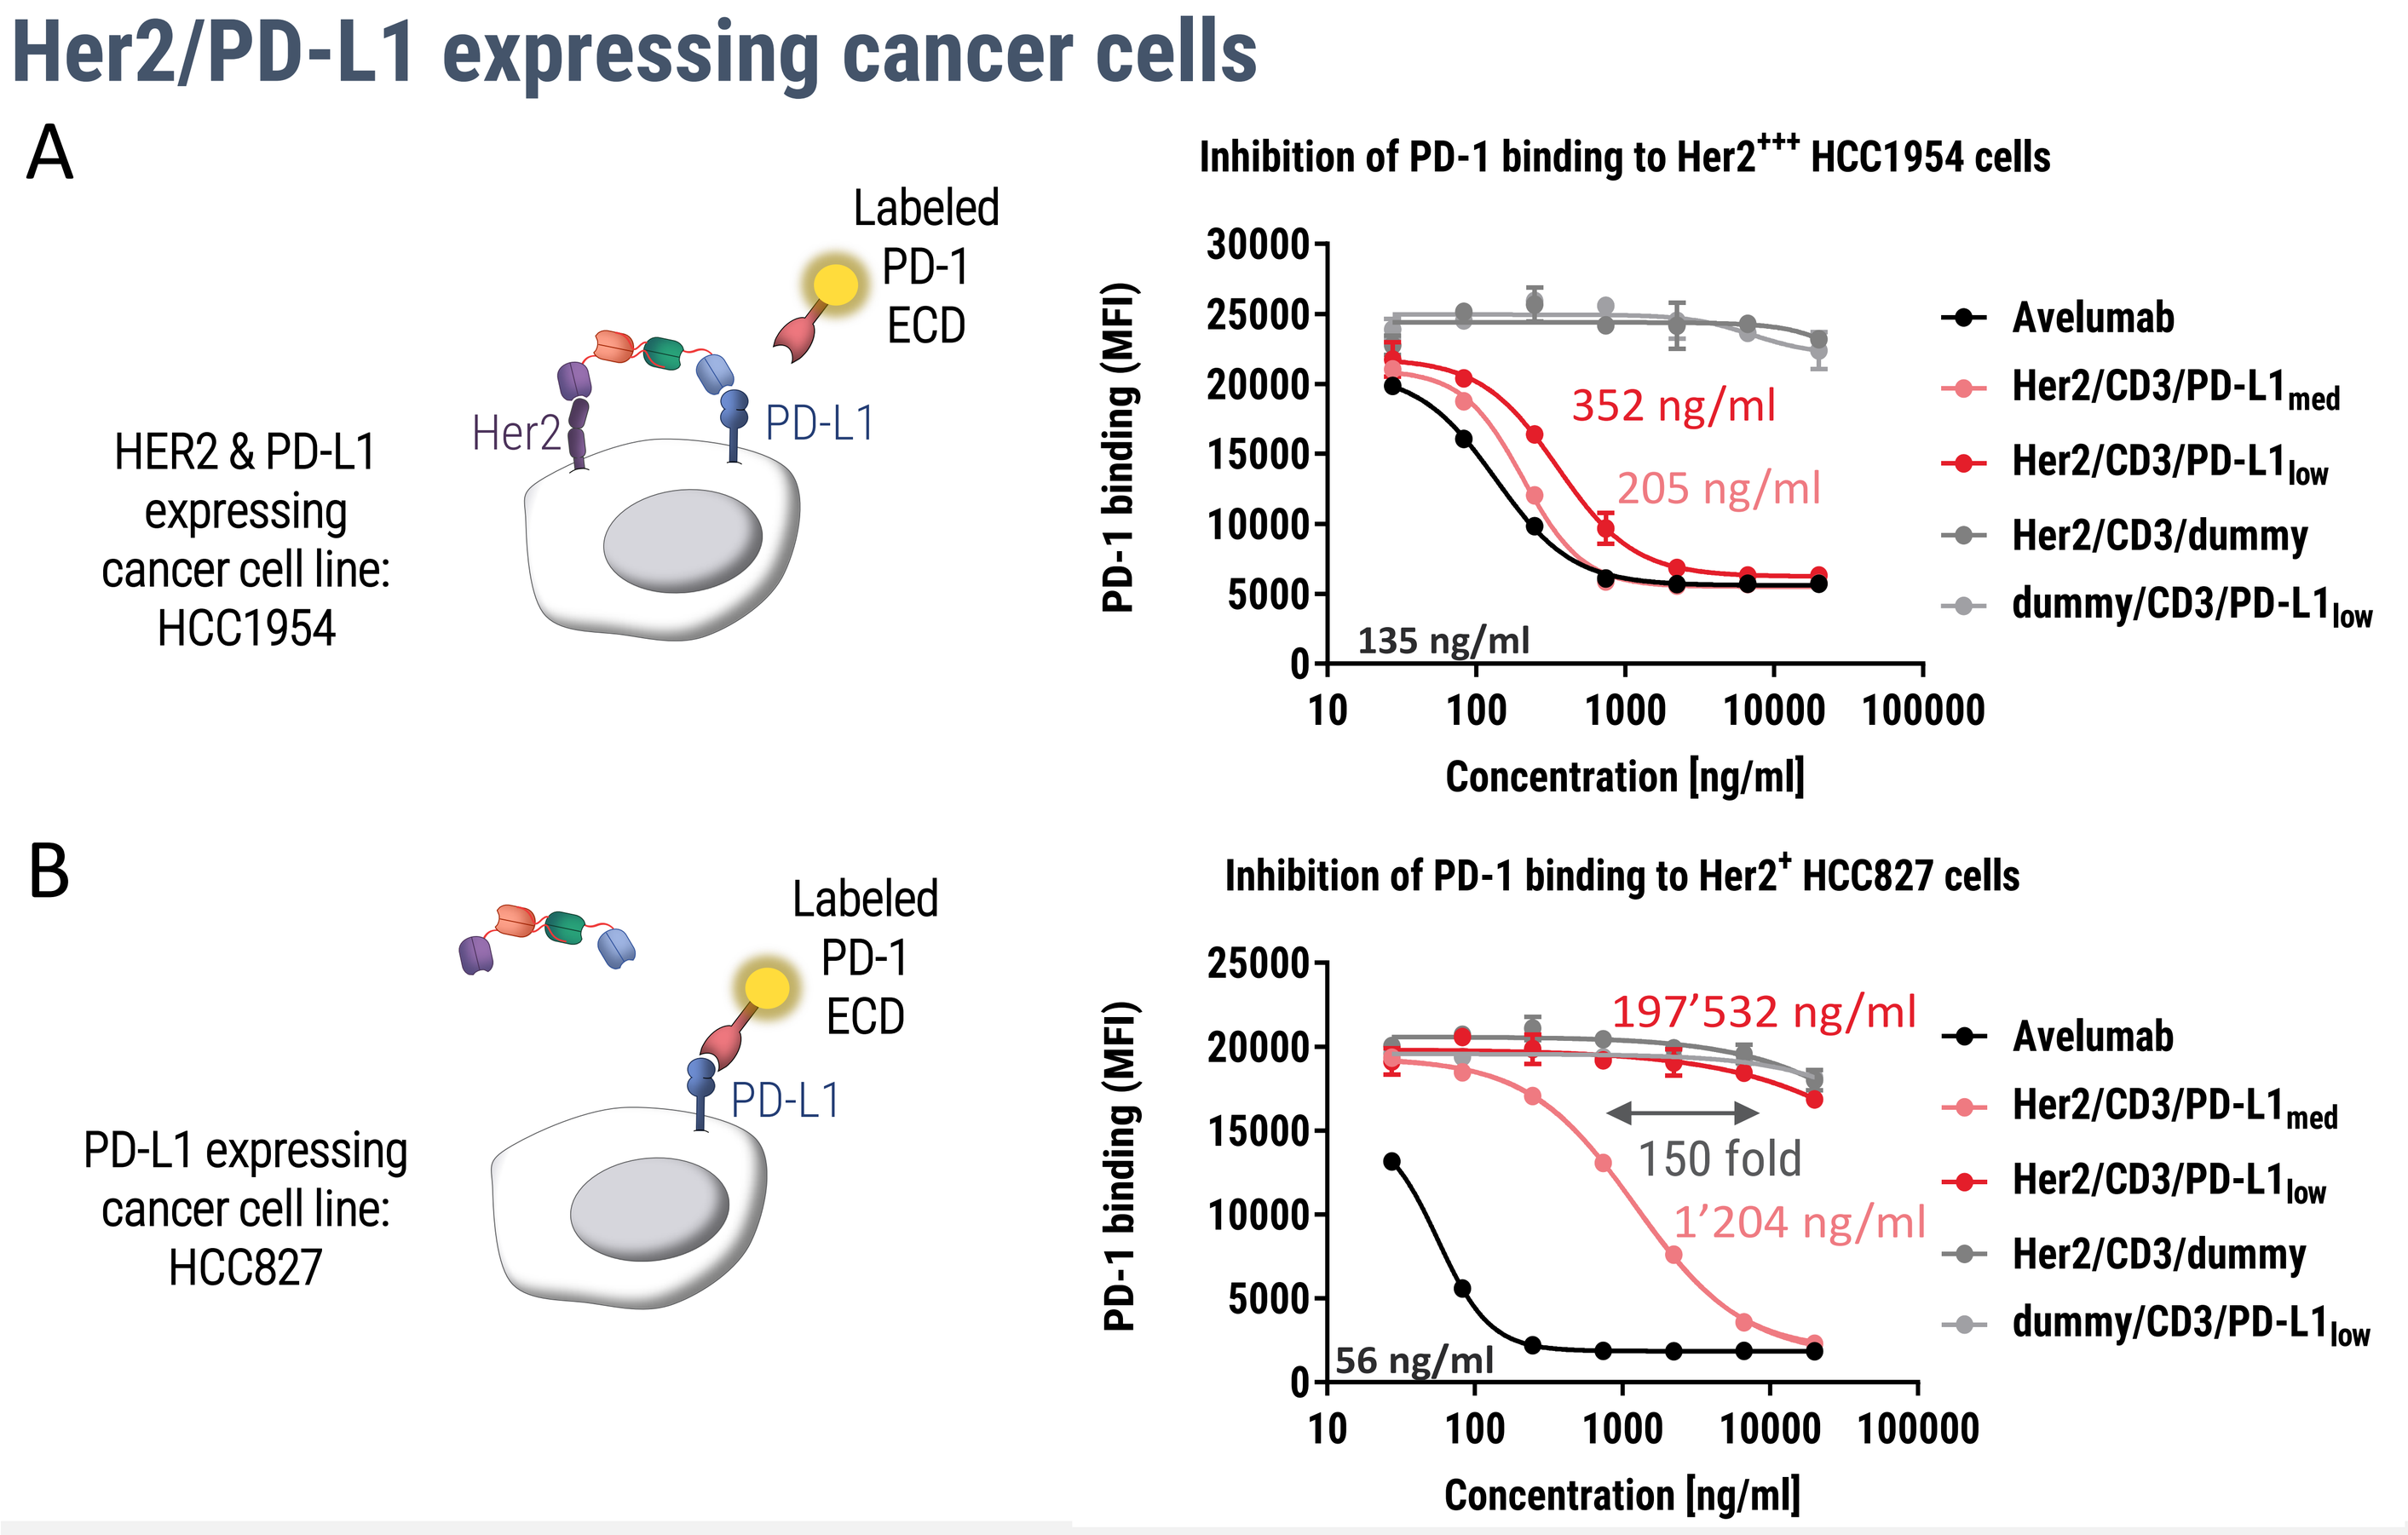

A
HER2 & PD-L1 expressing cancer cell line: HCC1954
Labeled PD-1 ECD
Inhibition of PD-1 binding to Her2+++ HCC1954 cells
352 ng/ml
205 ng/ml
135 ng/ml
PD-1 binding (MFI)
Concentration [ng/ml]
Avelumab
Her2/CD3/PD-L1med
Her2/CD3/PD-L1low
Her2/CD3/dummy
dummy/CD3/PD-L1low
B
PD-L1 expressing cancer cell line: HCC827
Inhibition of PD-1 binding to Her2+ HCC827 cells
197'532 ng/ml
150 fold
1'204 ng/ml
56 ng/ml

Figure 2. Neutralization of PD-1-binding to PD-L1$^+$ cells. Effect of PD-L1-affinity on potency of blockade of PD-1-binding to **A)** Her2^{+++}PD-L1$^+$ HCC1954 ductal breast carcinoma cells and **B)** Her2$^+$PD-L1$^+$ HCC827 NSCLC cancer cells. **PD-L1-blockade is restricted to high Her2 expressing cancer cells. Careful selection of the affinity of the αPD-L1 domain (10'000-fold lower than Avelumab) allows considerable extension of the therapeutic window.**

Her2/CD3/PD-L1$_{low}$/hSA tetra-specific MATCH4 molecule

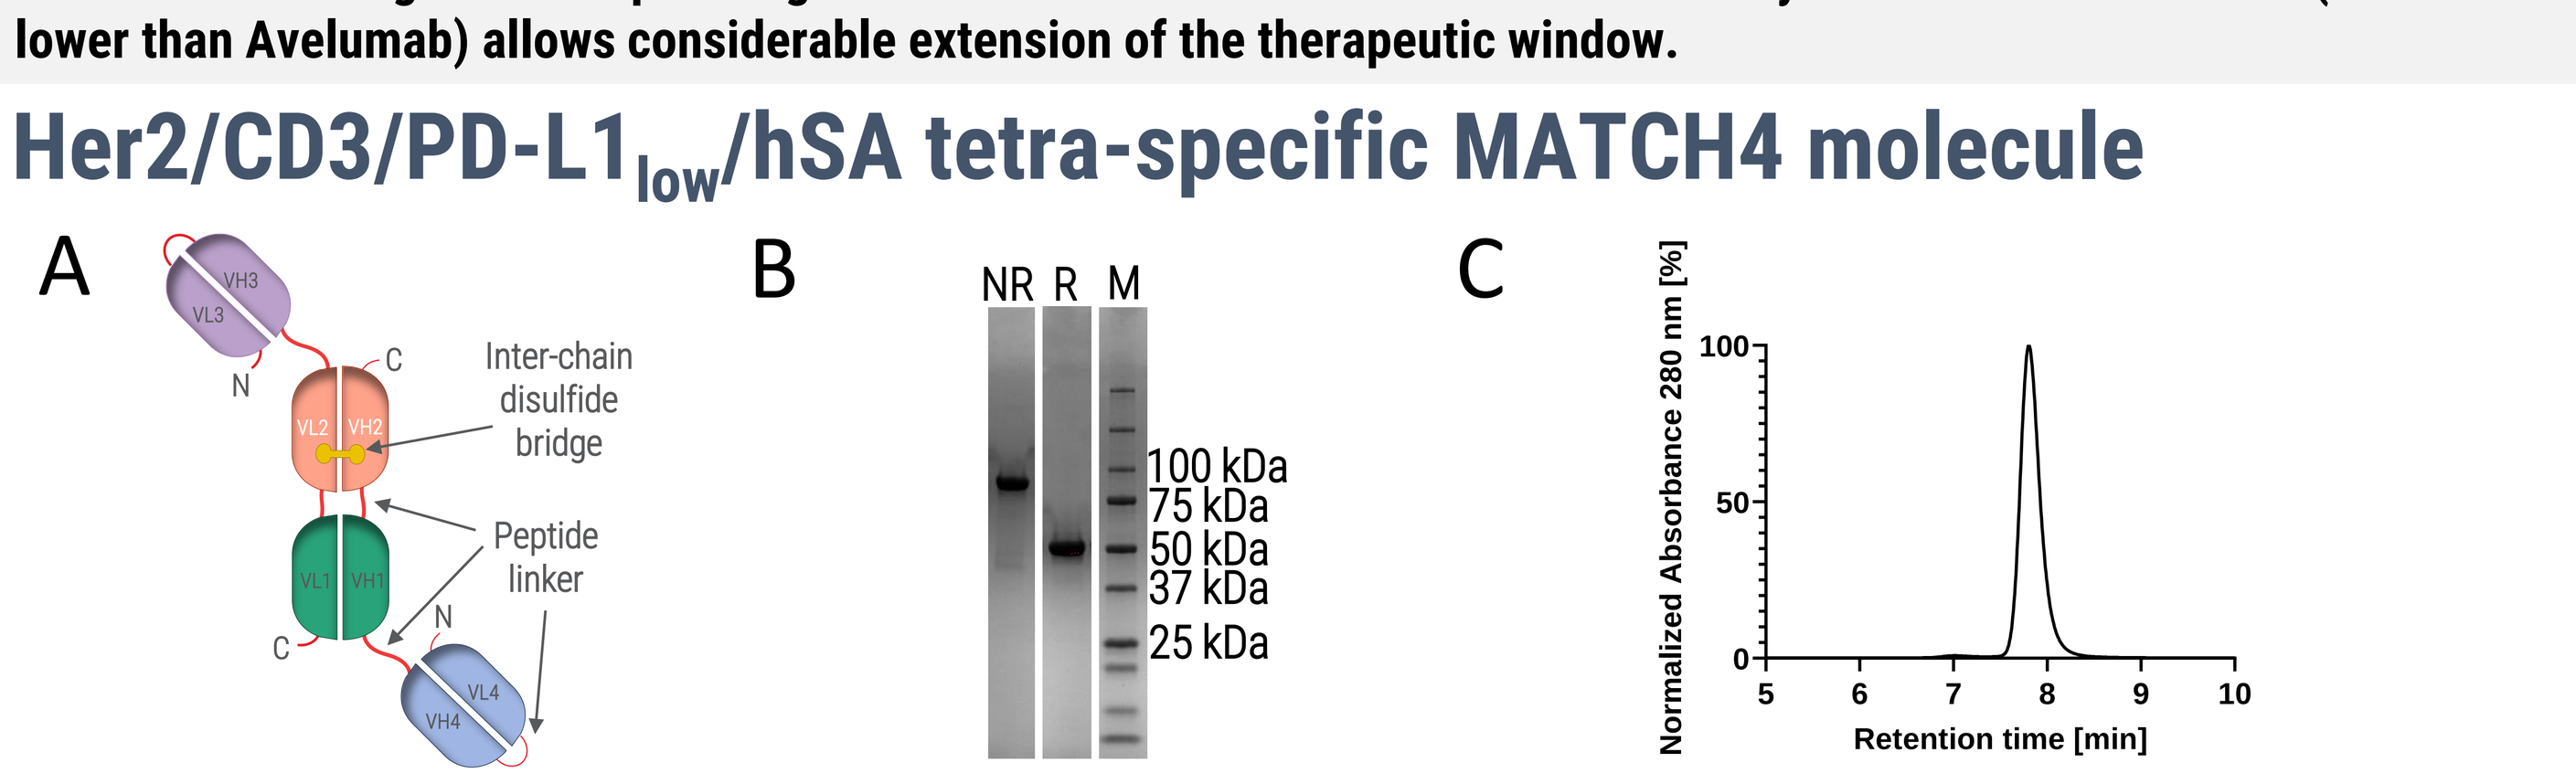

A
Inter-chain disulfide bridge
Peptide linker
B
NR R M
100 kDa
75 kDa
50 kDa
37 kDa
25 kDa
C
Normalized Absorbance 280 nm [%]
Retention time [min]

Figure 3. A) Schematic representation of Her2/CD3/PD-L1$_{low}$/hSA tetra-specific MATCH4 molecule: αCD3 (VL1/VH1 in green), αhSA (VL2/VH2 in orange), αHer2 (VL3/VH3 in purple), αPD-L1 (VL4/VH4 in blue), inter-chain disulfide bridge indicated in yellow. Domains are connected by Gly-Ser peptide linkers, as indicated by red lines. **B)** SDS-PAGE analysis of the MATCH4, NR: non-reducing, R: reducing, M: molecular weight marker **C)** Representative SE-HPLC chromatogram of the MATCH4.

Tumor targeted PD-L1 blockade

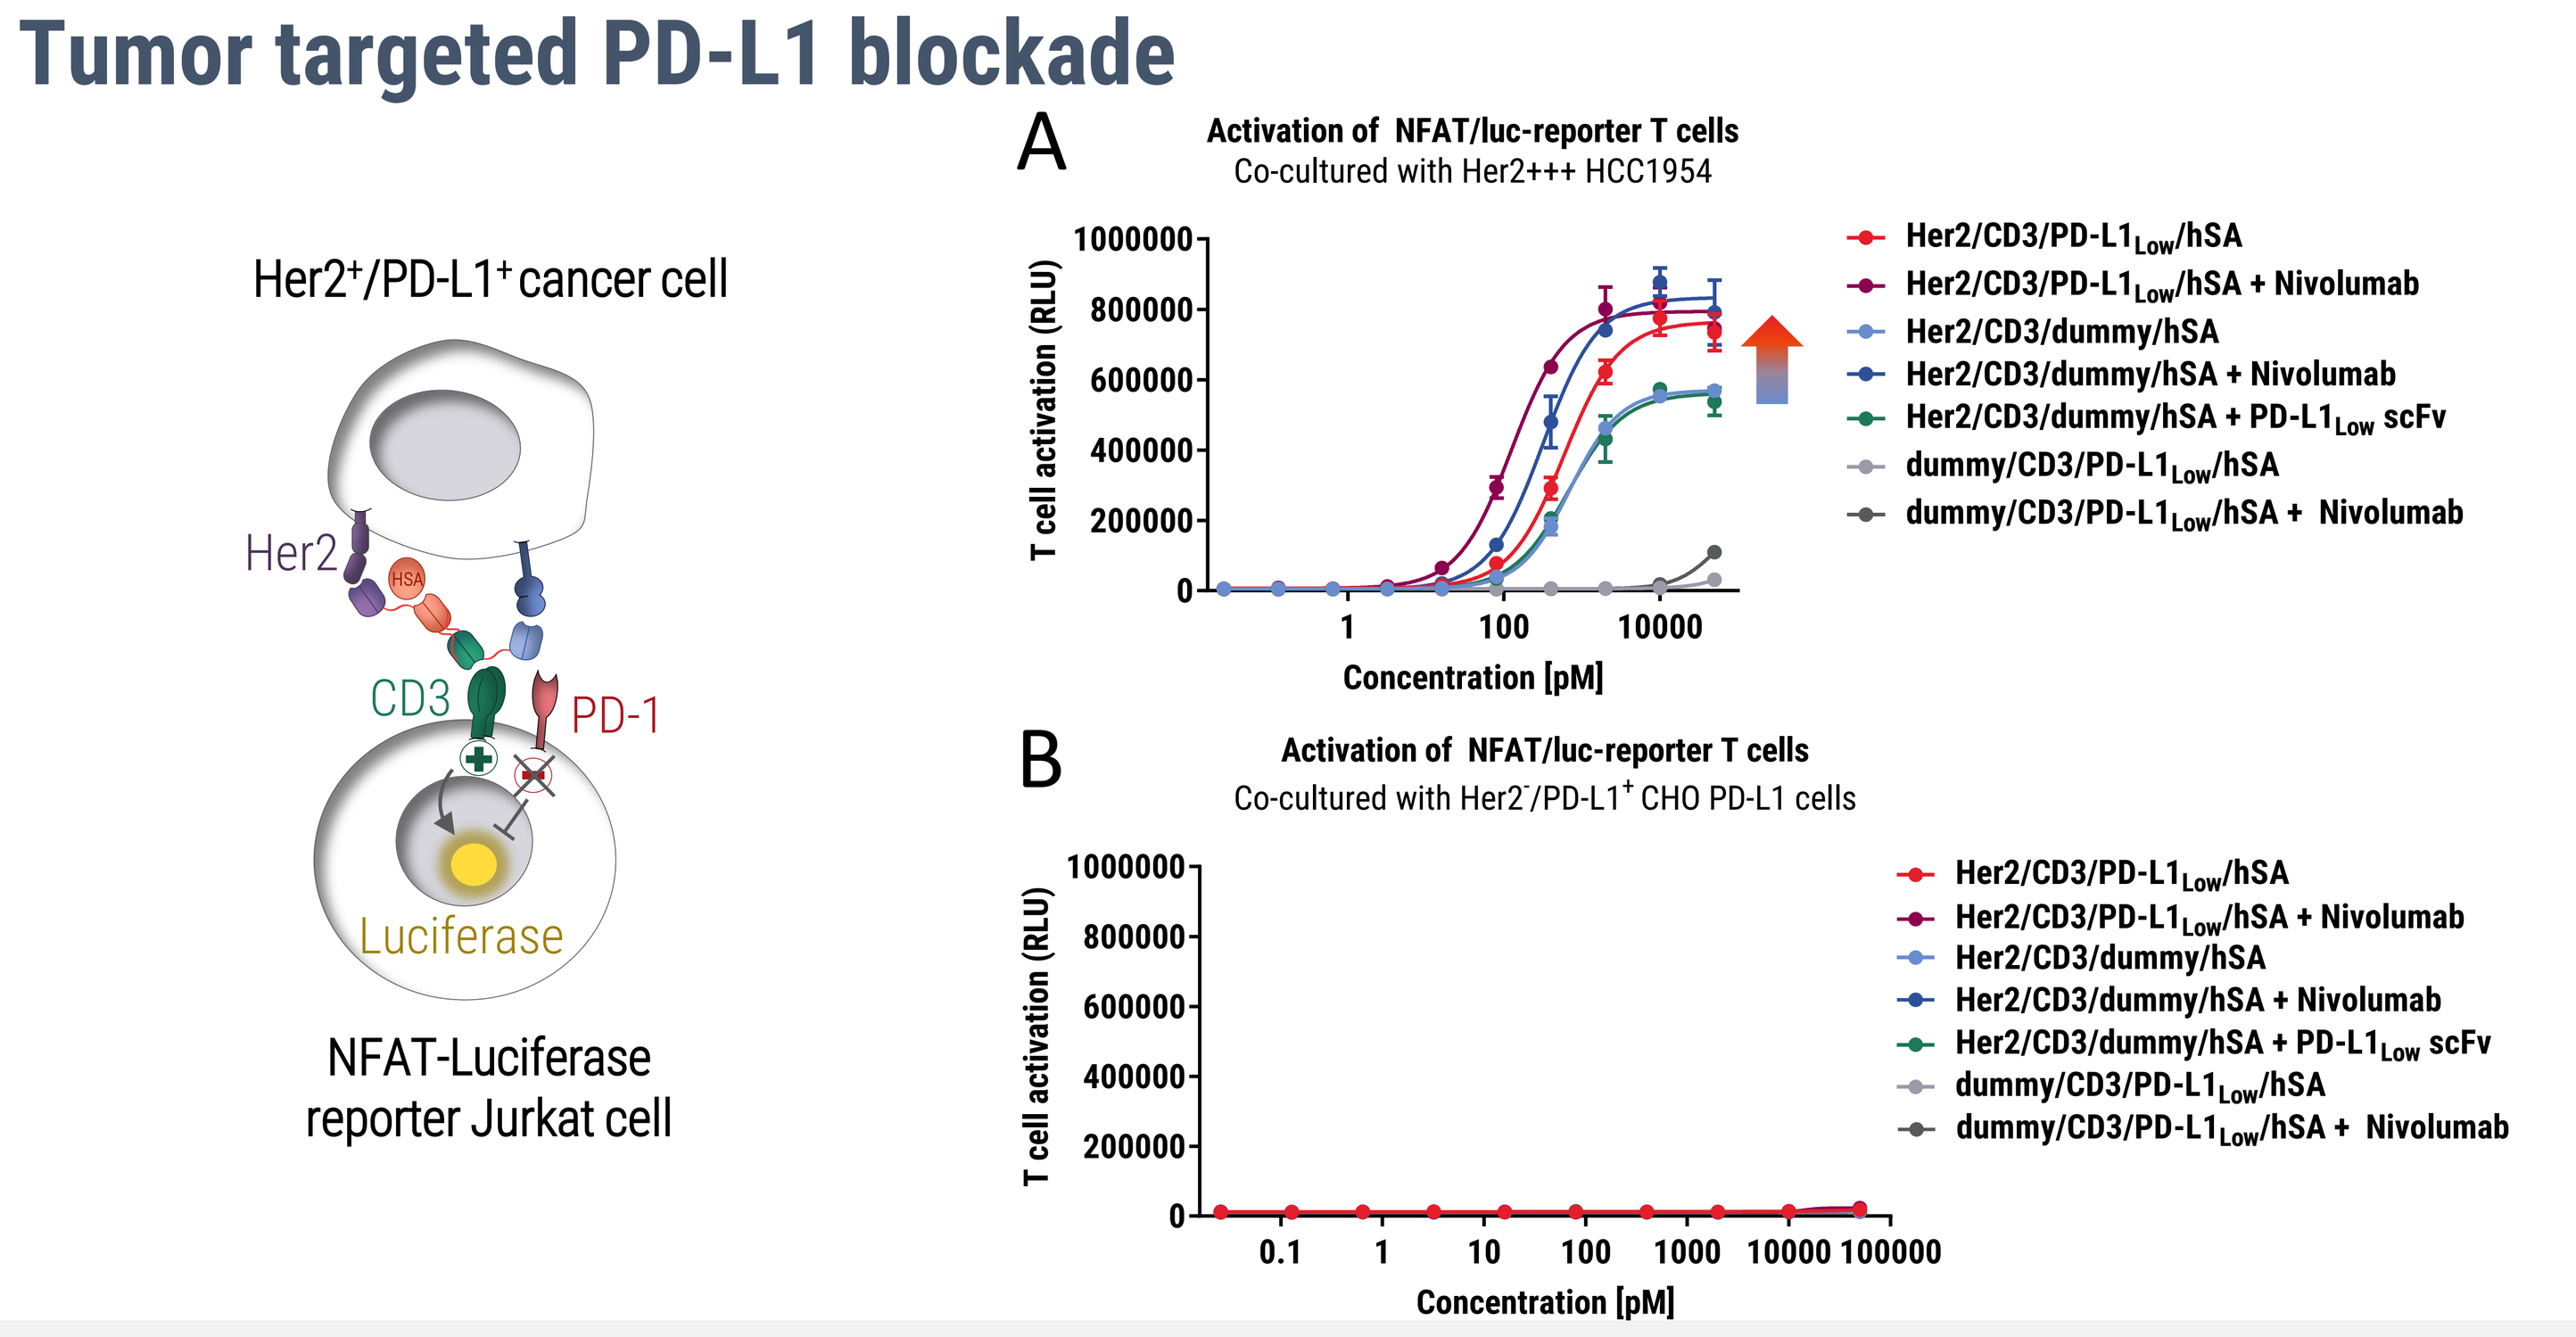

Her2+/PD-L1+ cancer cell
NFAT-Luciferase reporter Jurkat cell
Luciferase
A
Activation of NFAT/luc-reporter T cells
Co-cultured with Her2+++ HCC1954
B
Activation of NFAT/luc-reporter T cells
Co-cultured with Her2-/PD-L1+ CHO PD-L1 cells
T cell activation (RLU)
Concentration [pM]
Her2/CD3/PD-L1Low/hSA
Her2/CD3/PD-L1Low/hSA + Nivolumab
Her2/CD3/dummy/hSA
Her2/CD3/dummy/hSA + Nivolumab
Her2/CD3/dummy/hSA + PD-L1Low scFv
dummy/CD3/PD-L1Low/hSA
dummy/CD3/PD-L1Low/hSA + Nivolumab

Figure 4. Blockade of PD-L1-signaling in the immunological synapse. CD3-engagement and blockade of PD-1-signaling by MATCH4 molecules was assessed in a transgenic NFAT-Luciferase reporter Jurkat cell-line. PD-1-signaling is induced by co-cultivation of PD-1$^+$ reporter Jurkat cells with **A)** Her2^{+++}PD-L1$^+$ HCC1954 cancer cells or **B)** Her2−PD-L1$^+$ CHO cells. CD3 activation results in reporter gene activation and PD-L1-inhibition enhances maximum T cell-activation. **CD3 activation requires the presence of Her2$^+$ cells, and immunological synapse-restricted PD-L1-blockade elevates the efficacy of T cell-activation similarly to Nivolumab. When co-administered as an scFv, the low-affinity αPD-L1 domain has no effect.**

αPD-L1 binding-domain substantially increases potency

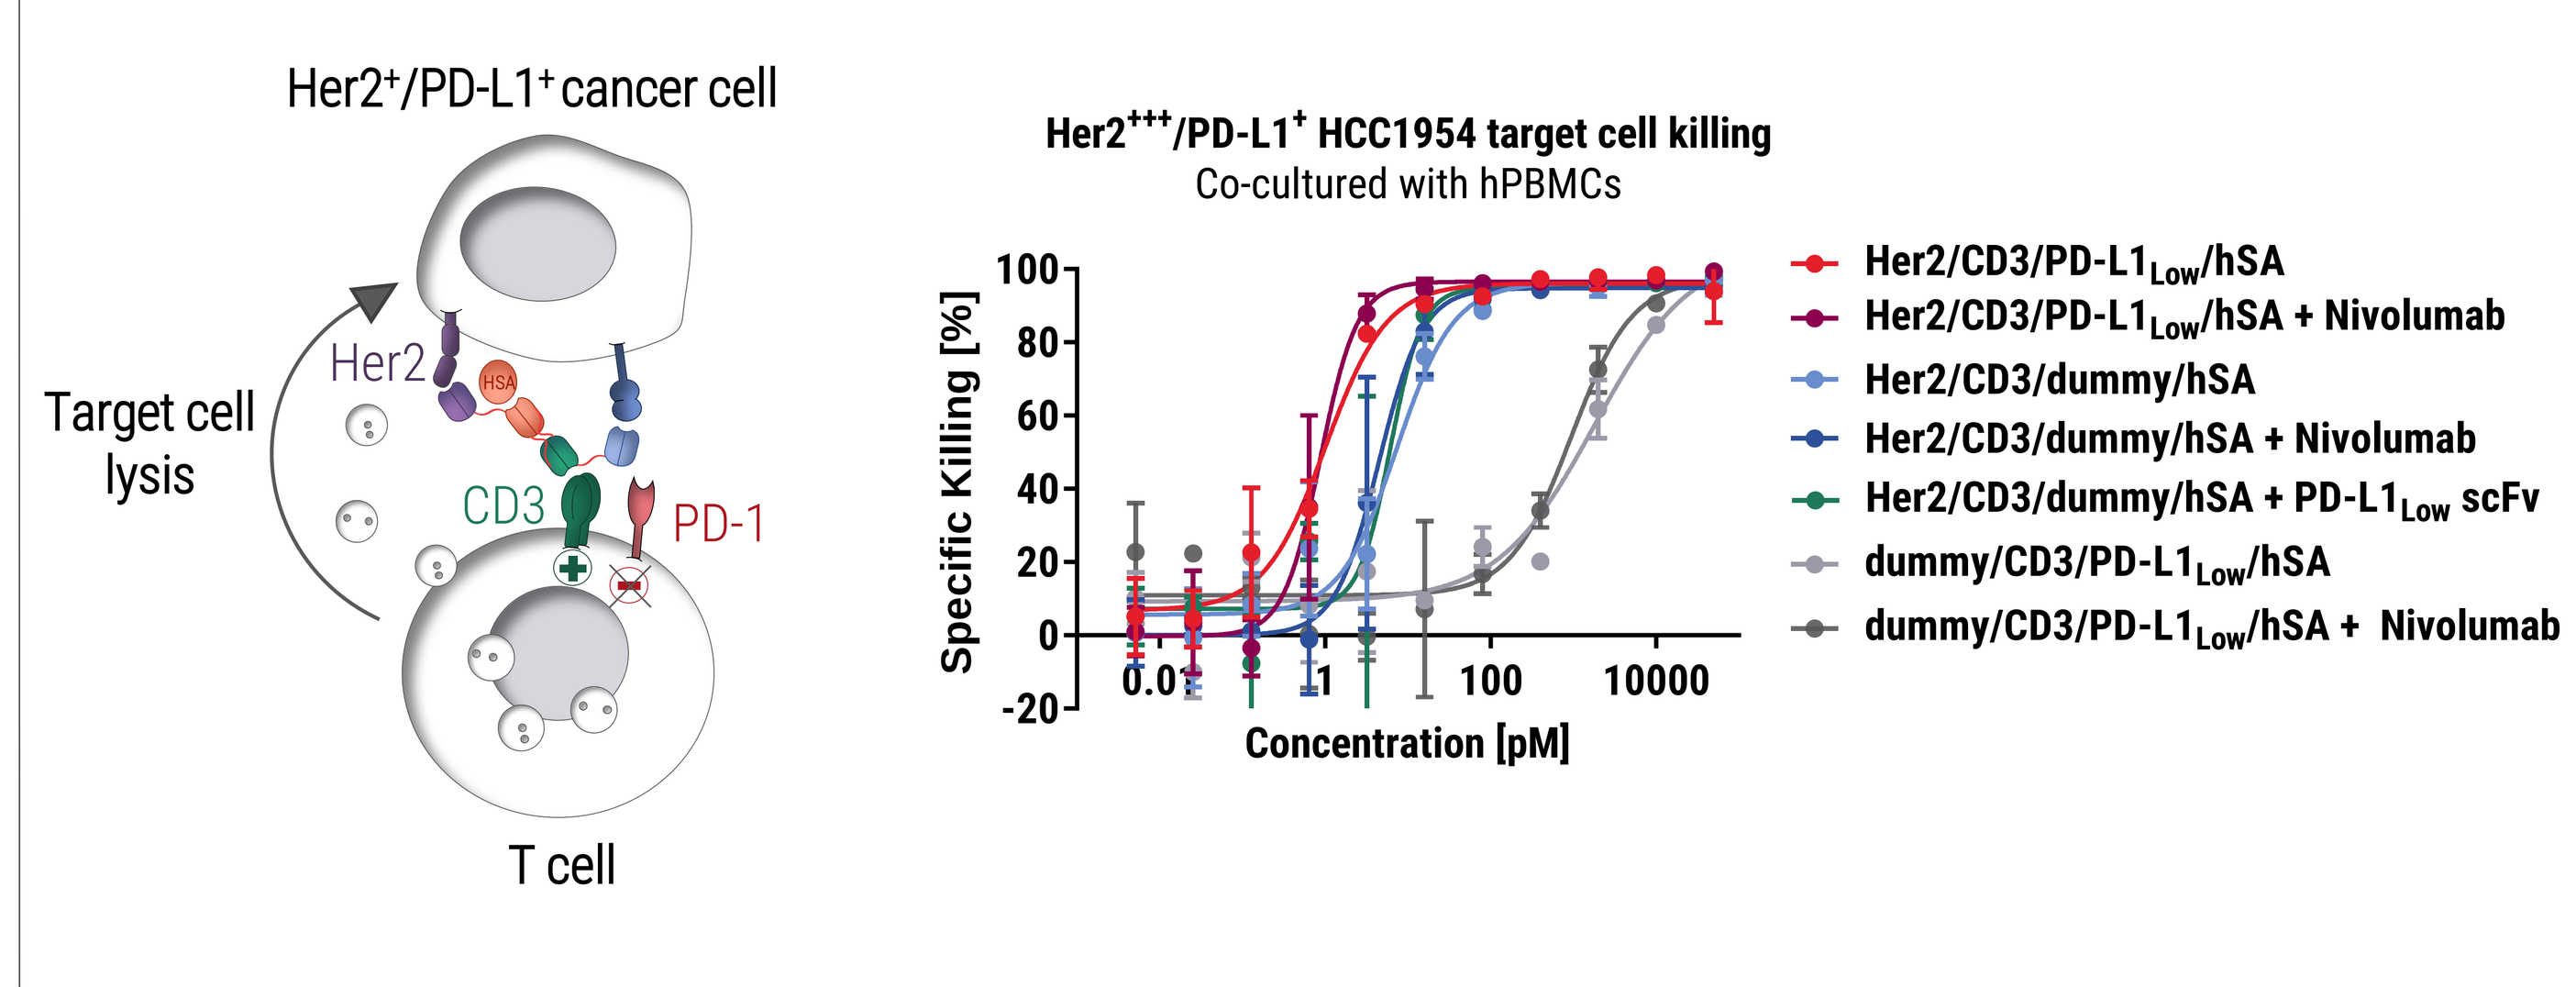

Her2+/PD-L1+ cancer cell
Target cell lysis
T cell
Her2+++/PD-L1+ HCC1954 target cell killing
Co-cultured with hPBMCs
Specific Killing [%]
Concentration [pM]
Her2/CD3/PD-L1Low/hSA
Her2/CD3/PD-L1Low/hSA + Nivolumab
Her2/CD3/dummy/hSA
Her2/CD3/dummy/hSA + Nivolumab
Her2/CD3/dummy/hSA + PD-L1Low scFv
dummy/CD3/PD-L1Low/hSA
dummy/CD3/PD-L1Low/hSA + Nivolumab

Figure 5. T cell-mediated target cell-killing. Freshly isolated human PBMCs were co-cultured for 40h with Her2^{+++}PD-L1$^+$ HCC1954 cells in the presence of the different molecules and target cell-killing was assessed by annexin V staining. MATCH4 molecules were tested in the presence or absence of a saturating concentration of Nivolumab (1 µg/ml). **MATCH4 harboring the low-affinity αPD-L1 domain is more potent than the MATCH 4 molecule lacking the αPD-L1 domain. Absence of the αHer2 domain results in a 10000-fold lower potency. Nivolumab treatment, in combination with the Her2/CD3/dummy/hSA tetra-specific MATCH4 molecule, did not change potency of the molecule, providing evidence that the improved potency the Her2/CD3/PD-L1$_{LOW}$/hSA MATCH4 is exclusively due to high avidity conferred by concomitant binding to both PD-L1 and Her2.**

PD-L1 binding domain substantially increases molecule potency

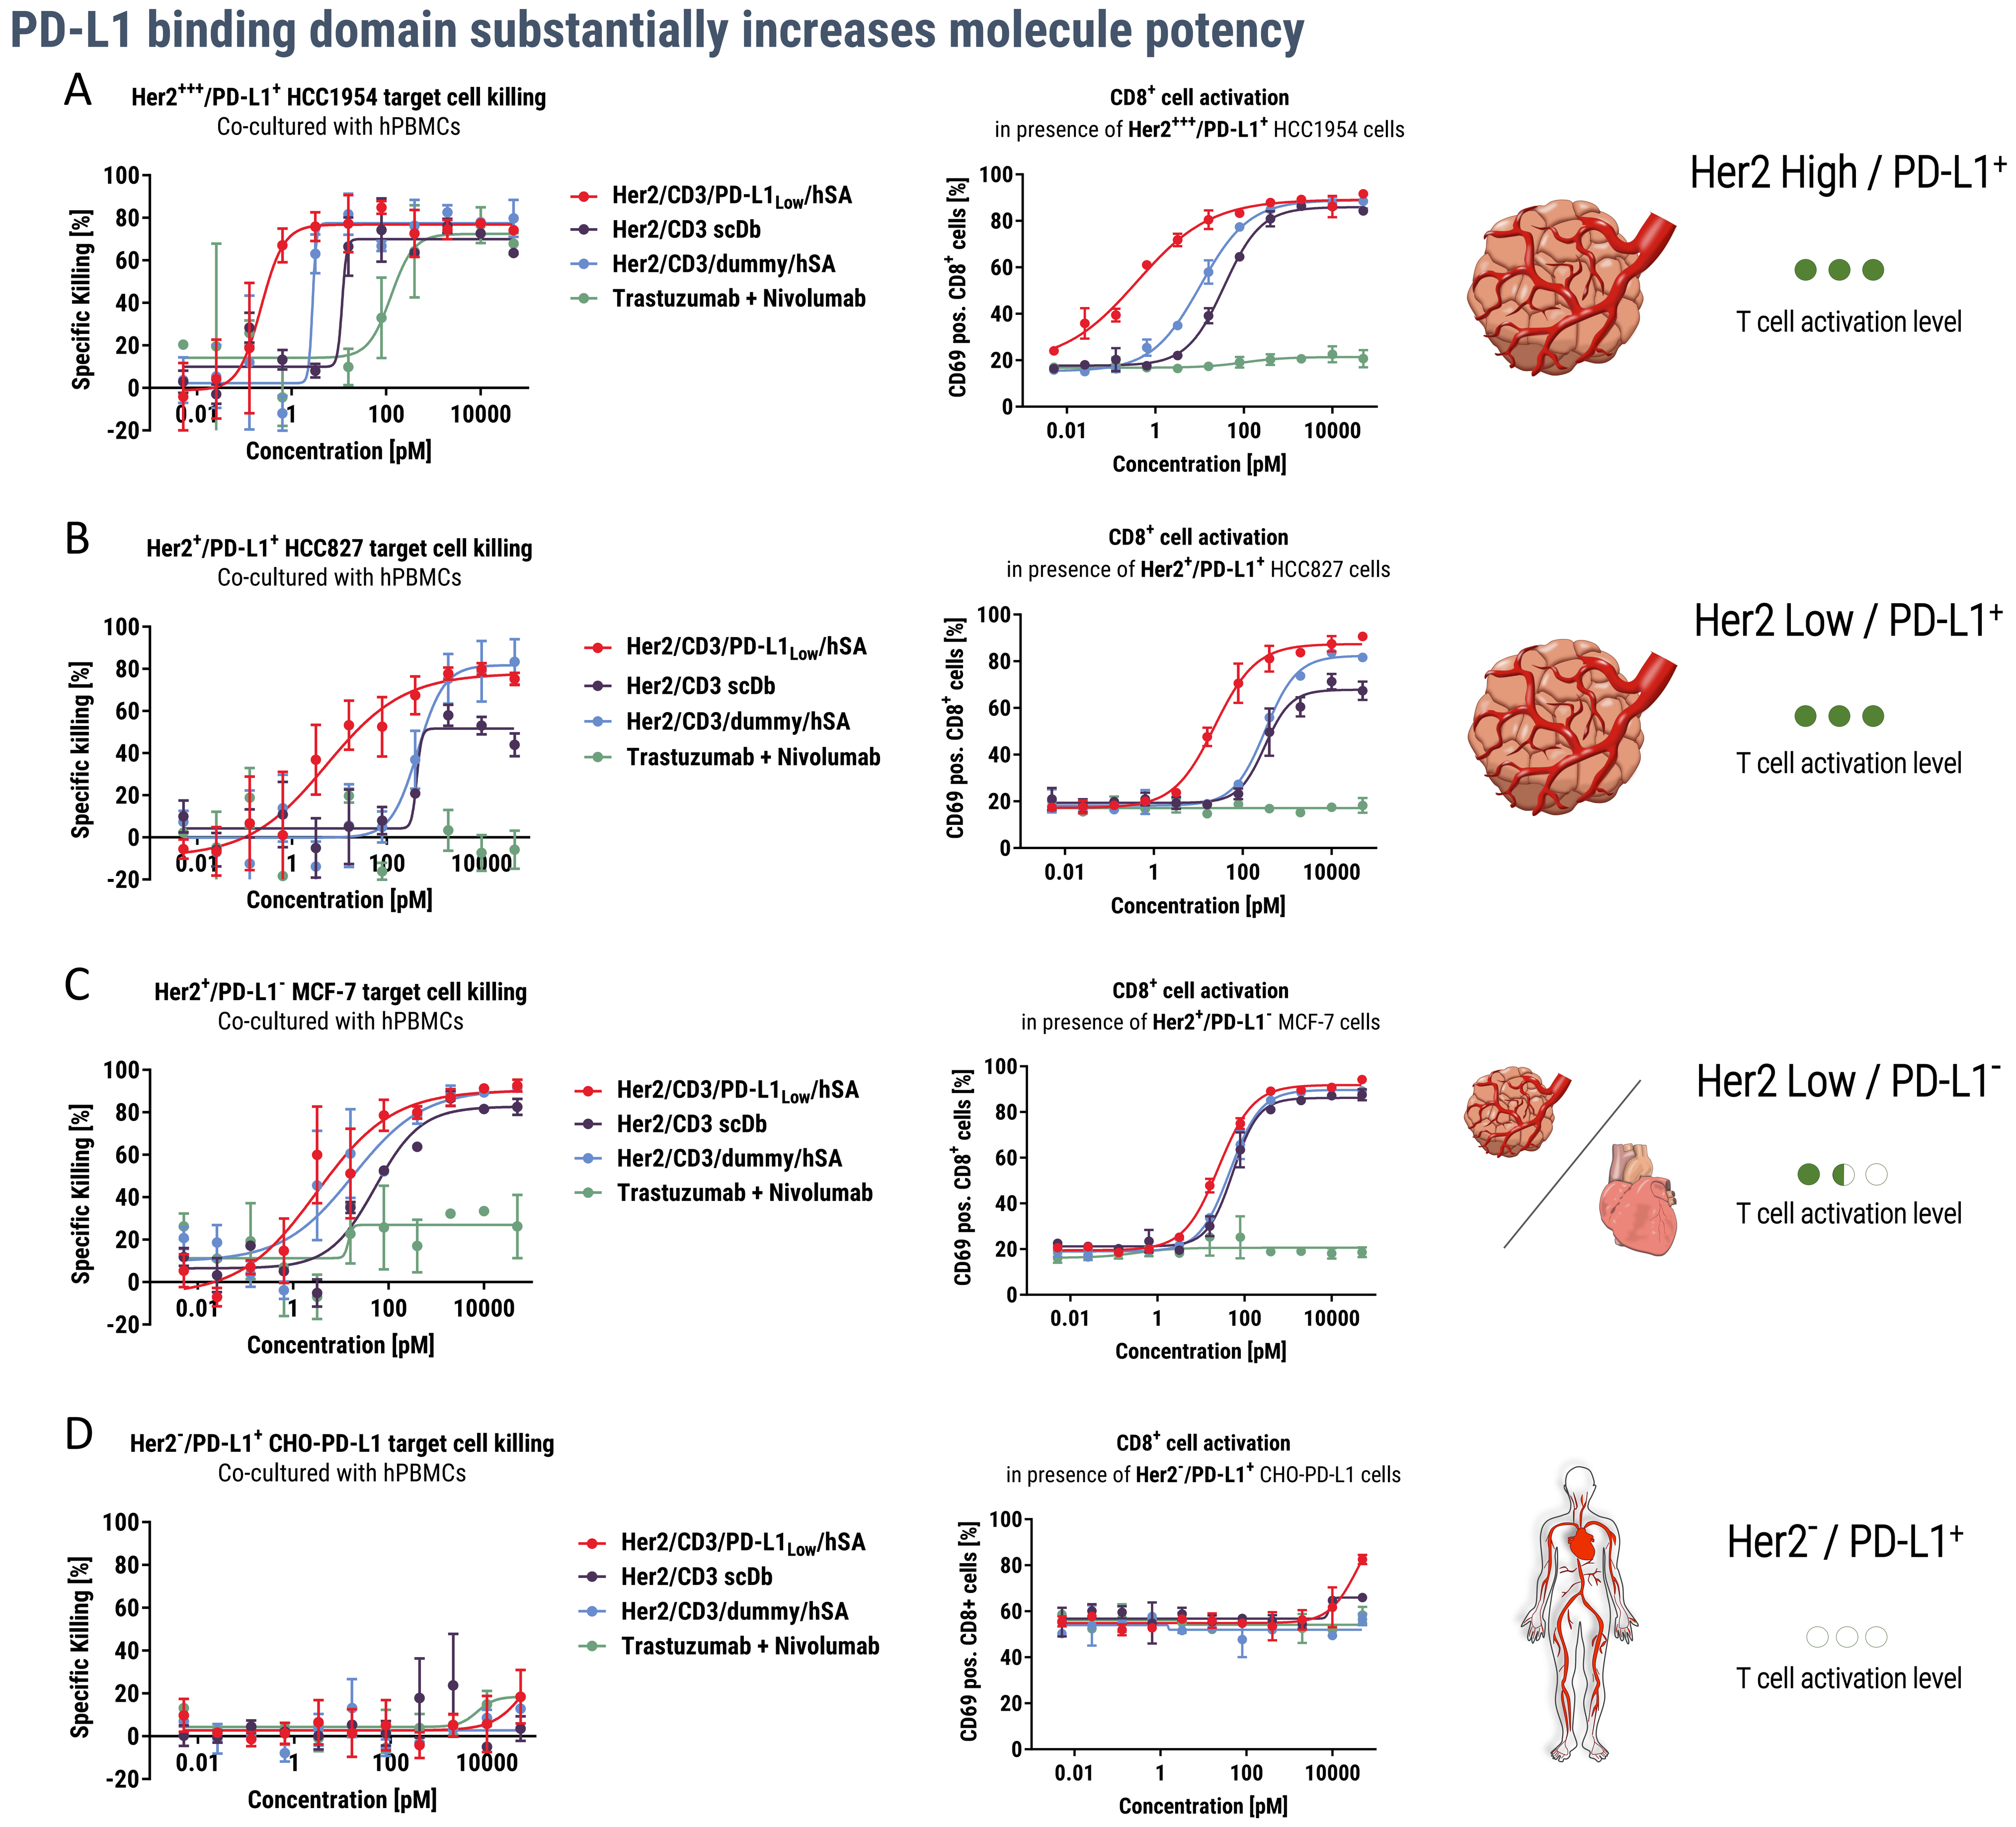

A
Her2+++/PD-L1+ HCC1954 target cell killing
Co-cultured with hPBMCs
CD8+ cell activation
in presence of Her2+++/PD-L1+ HCC1954 cells
Her2 High / PD-L1+
B
Her2+/PD-L1+ HCC827 target cell killing
CD8+ cell activation
in presence of Her2+/PD-L1+ HCC827 cells
Her2 Low / PD-L1+
C
Her2+/PD-L1- MCF-7 target cell killing
CD8+ cell activation
in presence of Her2+/PD-L1- MCF-7 cells
Her2 Low / PD-L1-
D
Her2-/PD-L1+ CHO-PD-L1 target cell killing
CD8+ cell activation
in presence of Her2-/PD-L1+ CHO-PD-L1 cells
Her2- / PD-L1+
T cell activation level
Specific Killing [%]
CD69 pos. CD8+ cells [%]
Concentration [pM]
Her2/CD3/PD-L1Low/hSA
Her2/CD3 scDb
Her2/CD3/dummy/hSA
Trastuzumab + Nivolumab

Figure 6. T cell mediated target cell killing and CD8+ cell activation in presence of **A)** Her2^{+++}/PD-L1$^+$ HCC1954 ductal breast carcinoma cells, **B)** Her2$^+$/PD-L1$^+$ HCC827 NSCLC cells, **C)** Her2$^+$/PD-L1$^-$ MCF-7 breast adenocarcinoma cells, **D)** Her2$^-$/PD-L1$^+$ CHO PD-L1 cells. Freshly isolated human PBMCs were co-cultured for 16h with indicated target cells in presence of the different molecules. **The MATCH4 Her2/CD3/PD-L1$_{low}$/hSA showed an increased potency in target cell killing and T cell activation compared to the Her2/CD3 scDb on Her2^{+++}/PD-L1$^+$ cells and Her2$^+$/PD-L1$^+$ cells. Activity of the MATCH4 molecule is similar to that of the Her2/CD3 scDb in the presence of Her2$^+$/PD-L1$^-$ cells. Importantly, no killing activity or T cell activation is observed in the presence of Her2$^-$/PDL1$^+$ cells.**

A

cell lines	Potency for T cell activation EC$_{50}$ (pM)	
	MATCH4	scDb
HCC1954	0.39	35.91
HCC827	23.72	314.6
MCF-7	25.33	54.29
CHO-PD-L1	NC	NC
Fold potency difference Her2 high / PDL1$^+$ vs Her2 low / PDL1$^-$	65x	1.5x

B

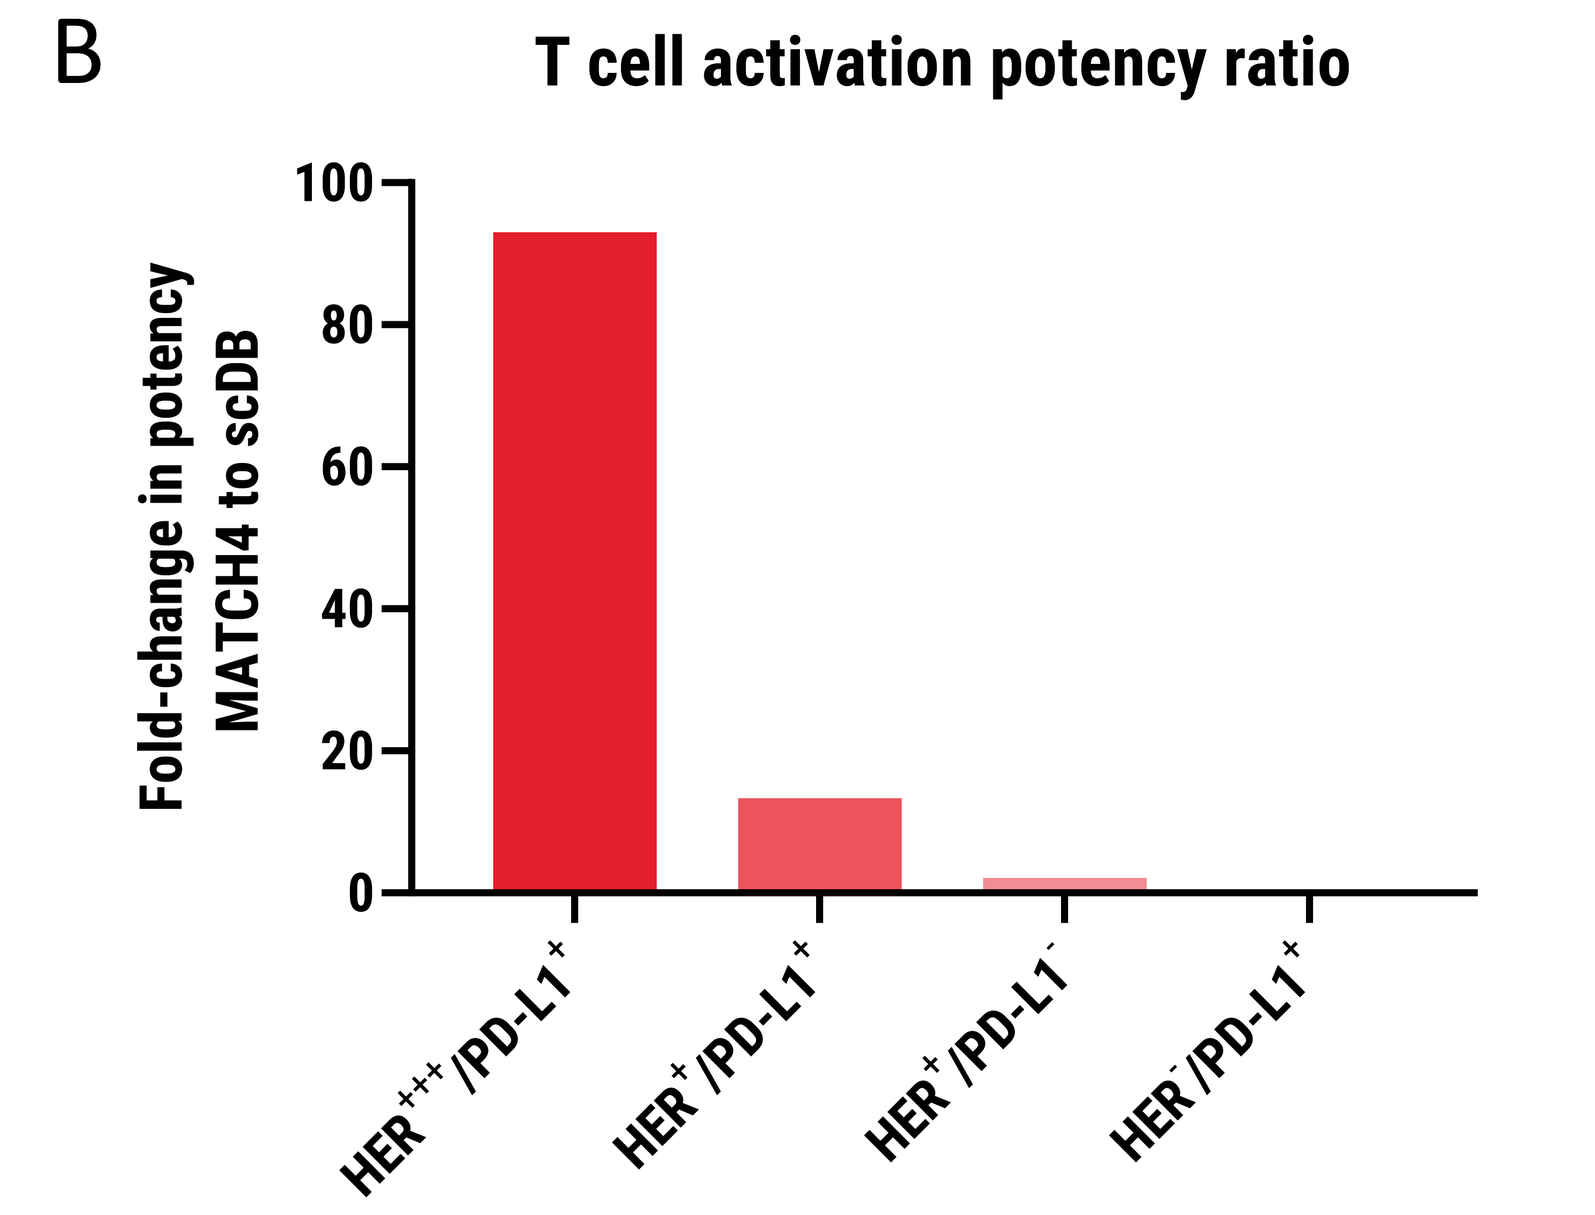

T cell activation potency ratio
Fold-change in potency MATCH4 to scDB
HER+++/PD-L1+
HER+/PD-L1+
HER+/PD-L1-
HER-/PD-L1+

C

cell lines	Her2 expression Antibody binding sites
HCC1954	61937
MCF-7	12670
HCC827	8908

Figure 7. T cell activation potencies of MATCH4 and scDb on cells expressing different Her2 and PD-L1 levels. **A)** EC50 values for T cell activation mediated by MATCH4 and scDb molecules. **B)** Calculated potency ratios between MATCH4 and scDb. **C)** Her2 antibody binding sites on different cancer cells. **The MATCH4 Her2/CD3/PD-L1$_{low}$/hSA showed an increased potency on PD-L1 positive Her2 expressing cancer cells in comparison to the scDb regardless of Her2 expression levels. Data suggest an improved therapeutic window for the MATCH4 over the scDb in the context of PD-L1 expression. Importantly a greater difference in potency can be demonstrated for the MATCH4 molecule between Her2 high / PD-L1$^+$ cells and Her2 low / PD-L1$^-$ cells compared to a scDb. NC = not calculable**

Efficient tumor regression with the MATCH4 molecule

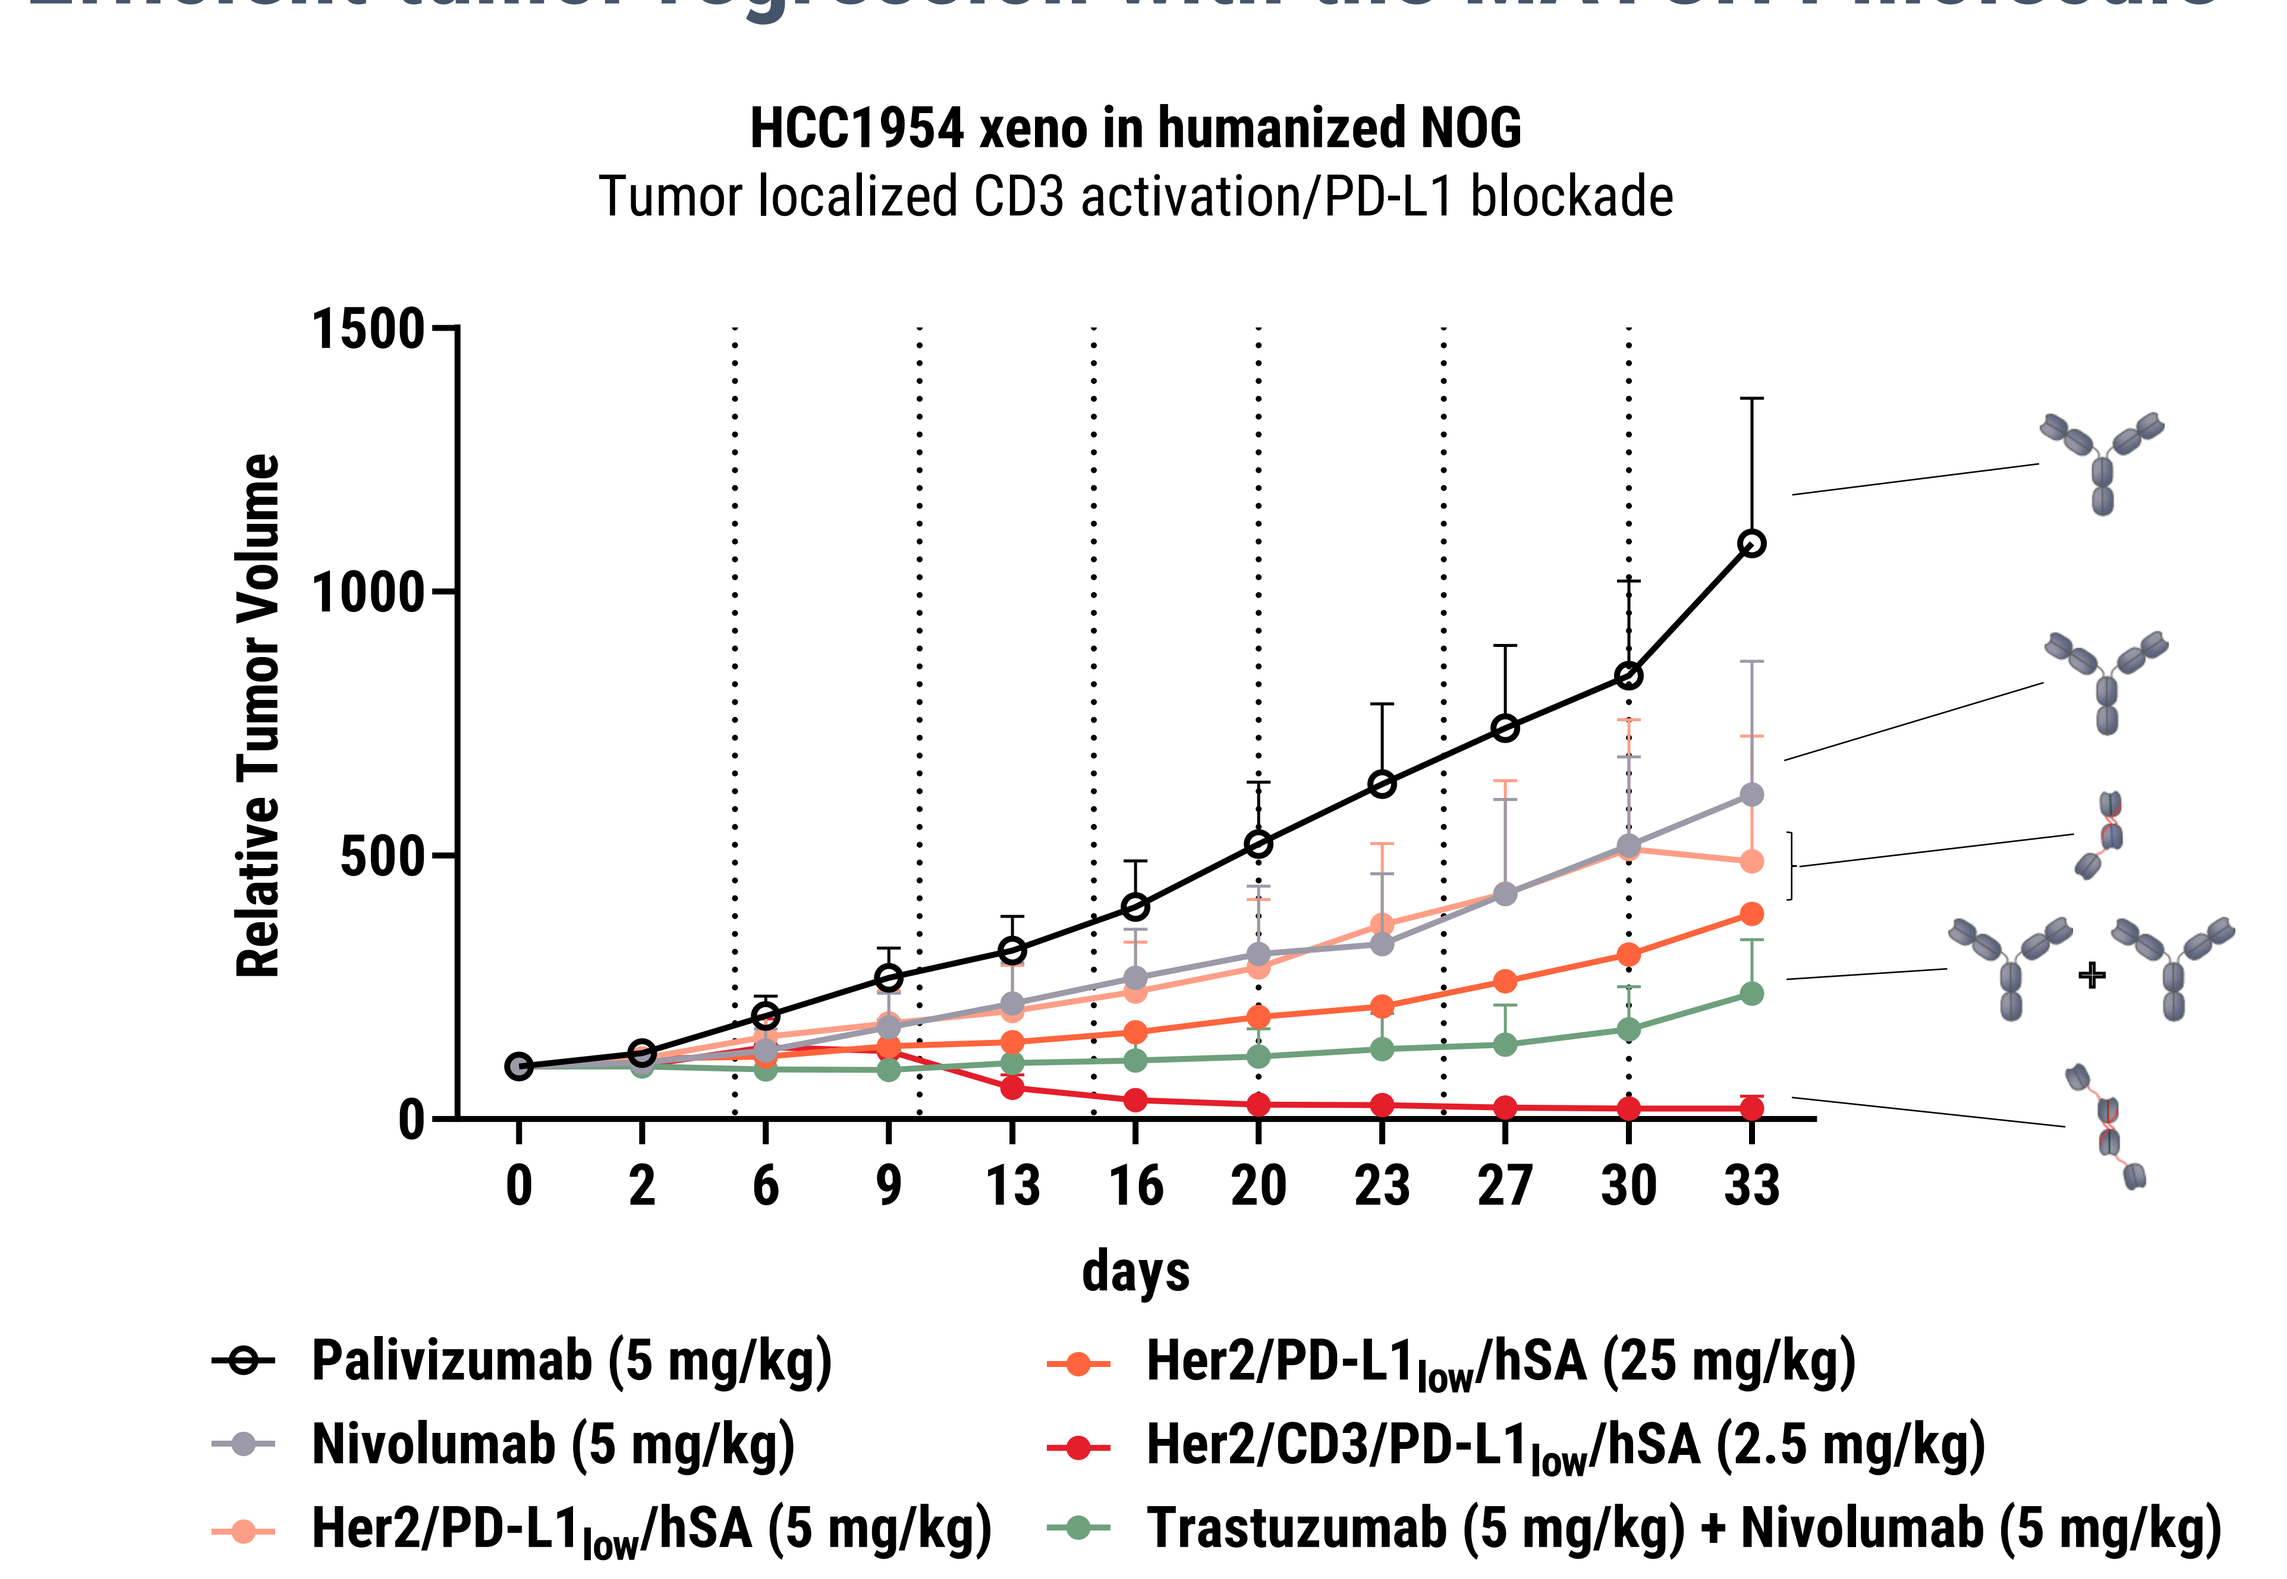

HCC1954 xeno in humanized NOG
Tumor localized CD3 activation/PD-L1 blockade
Relative Tumor Volume
days
Palivizumab (5 mg/kg)
Nivolumab (5 mg/kg)
Her2/PD-L1low/hSA (5 mg/kg)
Her2/PD-L1low/hSA (25 mg/kg)
Her2/CD3/PD-L1low/hSA (2.5 mg/kg)
Trastuzumab (5 mg/kg) + Nivolumab (5 mg/kg)

Figure 8. Human PBMC-substituted NOG mice were engrafted with HCC1954 ductal breast carcinoma cells (n=8 per group). Mice were treated on day 0, 5, 10, 15, 20, 25 and 30 (dotted vertical lines). Tumor growth and body weight was recorded twice weekly. **The Her2/CD3/PD-L1$_{low}$/hSA tetra-specific MATCH4 molecule elicited considerable tumor regression in all treated mice. The MATCH4 molecule demonstrated greater anti-tumoral efficacy than the Nivolumab + Trastuzumab combination. Nivolumab shows limited anti-tumoral activity in this model and the control molecule Her2/PD-L1$_{low}$/hSA shows a similar anti-tumoral activity despite a 5-fold lower potency in vitro (data not shown), confirming effective tumor-targeted PD-L1 blockade. No body weight loss was observed (not shown).**

Conclusions:

- A next-generation MATCH4 tetra-specific (Her2/CD3/PD-L1$_{low}$/hSA) molecule displays exquisite anti-tumoral efficacy in Her2$^+$ cancer models
- Increased therapeutic potential on Her2$^+$/PD-L1$^+$ cells whilst not increasing activity on Her2$^+$/PD-L1$^-$ cells (Fig 6A-C)
- No T cell activity in the presence of Her2$^-$ PD-L1$^+$ cells due to the use of a low-affinity anti-PD-L1 domain (Fig. 6D)
- PD-L1 / PD-1 antagonism is dependent on Her2 expression and delivers an additive effect to T cell activation at concentrations >100pM (Fig. 4)
- The anti-tumoral activity is predominantly driven by an avidity-effect of engaging both Her2 and PD-L1
- **Potential to open the therapeutic window of Her2/CD3 activity by increasing selectivity towards Her2 PD-L1 double-positive cells and therefore may present a safer Her2 targeting T cell engager**

34th Annual Meeting of the Society for Immunotherapy of Cancer, 2019. National Harbor, Maryland, USA. Poster P779